#174

Evidence of Early Bone Response After Initiation of ERT in a 3-Year Old Patient With MPS VII

Lau HA[1], Parmar S[1], Ferraris M[1], Haller C[2], Agarwal S[2], Kakkis E[2]

[1]Division of Neurogenetics, Department of Neurology, NYU Langone Medical Center, New York, NY, [2]Ultragenyx Pharmaceutical Inc., Novato, CA

ABSTRACT

Skeletal involvement is a common feature in the Mucopolysaccaridoses (MPS) and includes short stature, dysostosis multiplex, joint pain, stiffness and/or laxity. Early enzyme replacement therapy (ERT) in the first few years of life may reduce or prevent MPS bone disease manifestations. Potential skeletal benefits of early intervention in MPS VII have not been described. We report an early marked response in bone biomarkers in a young child after a single IV dose of rhGUS (UX003), which is under development as a potential ERT for MPS VII.

A 3-year-old male with MPS VII characterized by short stature (< 5% height for age) participating in an open-label trial of rhGUS 4 mg/kg biweekly in patients < 5 years had a normal baseline alkaline phosphatase (ALP) of 186 U/L which rose significantly to 1633 U/L (reference range 104-345) two weeks after the first infusion. Bone-specific ALP was mildly elevated at 111 U/L (reference range 31-103), and Procollagen Type 1 N-terminal propeptide (P1NP) was significantly increased at 470 mcg/L (reference range 22-105) indicative of robust bone formation. A marker of bone resorption, C-telopeptide (CTX) was within normal limits. The patient was asymptomatic with no recent illnesses, injuries or fractures. Other laboratory values including liver and thyroid function tests were unremarkable.

In MPS glycosaminoglycans accumulate in the lysosomes of cells including osteoclasts, which disrupts the normal cycle of bone formation and remodeling leading to structurally and functionally abnormal bone.

In this young child with MPS VII, a marked increase in bone turnover markers, P1NP and ALP was observed after a single IV dose of rhGUS. Although preliminary, this observation suggests replacement of β-glucuronidase early in life may activate the normal bone growth cycle and potentially reduce the long term skeletal manifestations of the disease. Additional bone biomarker data may further inform this observation.

PATIENT GROWTH CHART

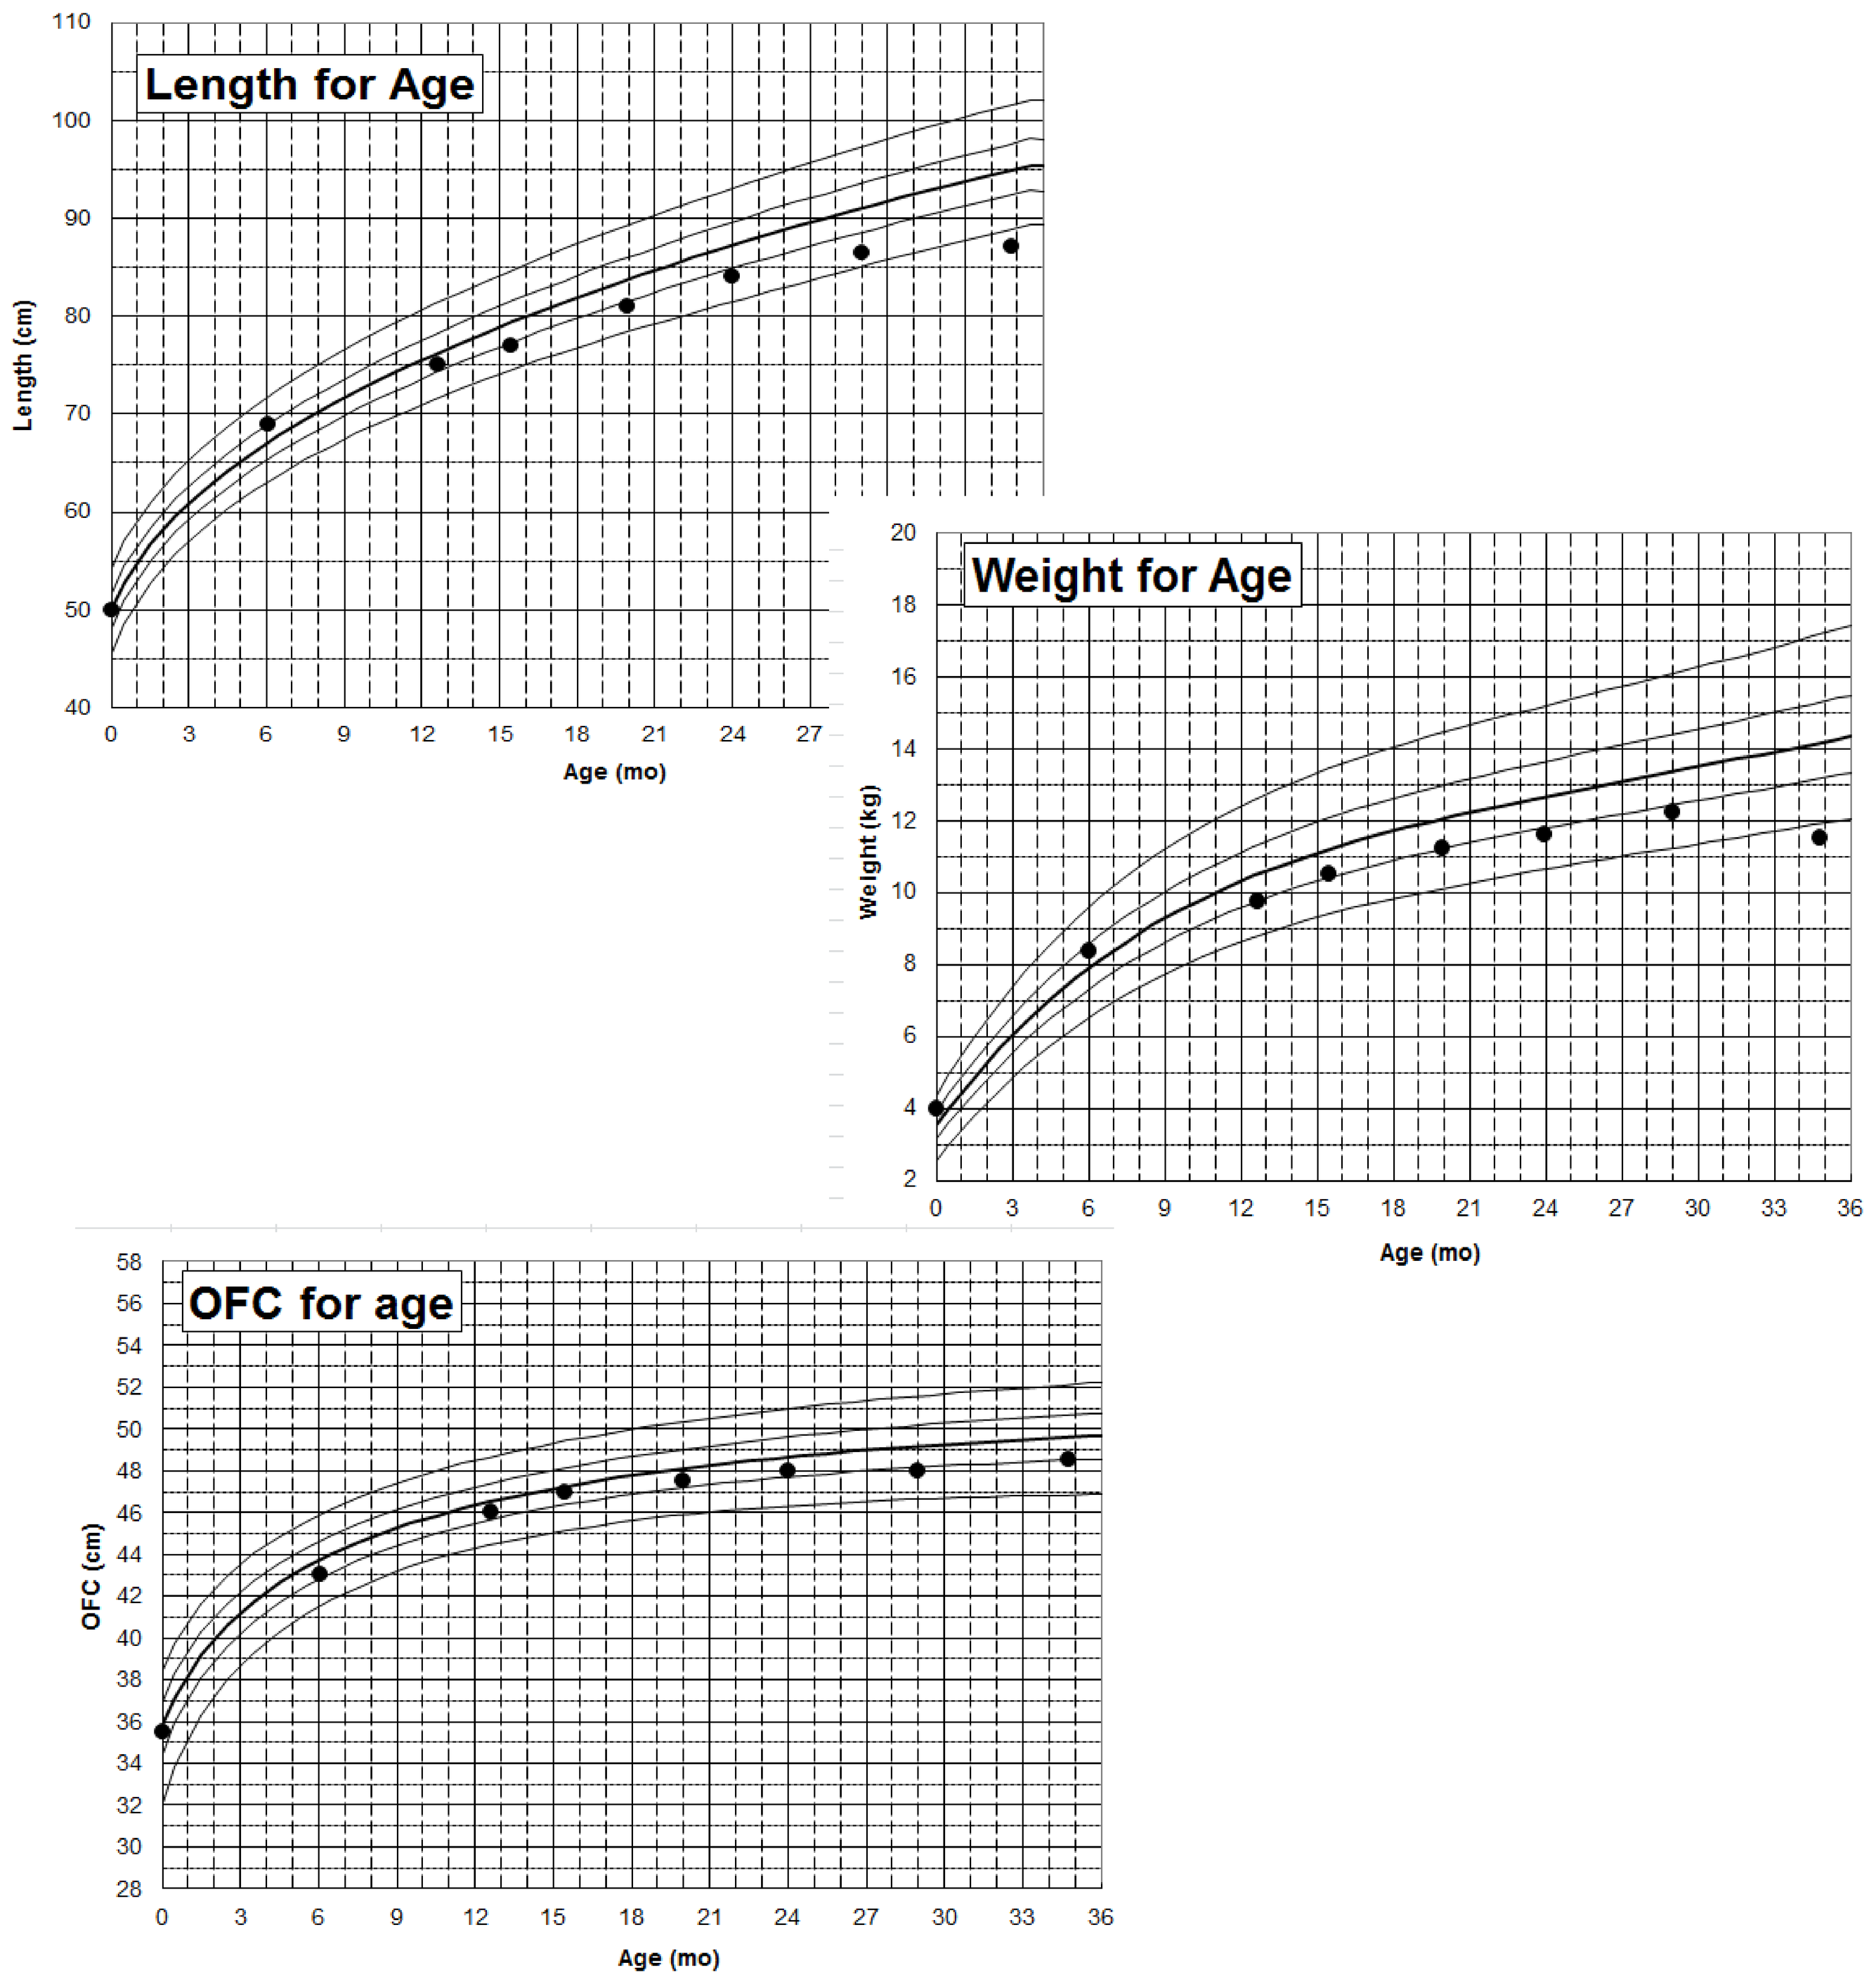

Length for Age
Length (cm)
Age (mo)
Weight for Age
Weight (kg)
Age (mo)
OFC for age
OFC (cm)
Age (mo)

Developed by Ernest M. Post, MD and Zimi Medical Technologies, LLC from CDC data on http://www.cdc.gov/growthcharts/

SUMMARY

- Alkaline Phosphatase (ALP)
 - ALP levels were normal at Baseline and increased by more than 4-fold the upper limit of the reference range by Week 2 after initiating UX003 therapy
 - ALP levels remained slightly elevated at Week 4 and then decreased to screening levels by Week 8
- Bone specific ALP (BALP) and Procollagen type 1 N-terminal propeptide (P1NP) are bone formation markers
 - At Week 4, BALP and P1NP were elevated indicating increased bone formation
 - At Week 16, BALP levels had returned to the reference range but P1NP remained substantially elevated
- C-Telopeptide (CTX) is a bone resorption biomarker
 - CTX levels were within normal limits for healthy children (Herrmann et al. 2014) suggesting bone resorption was not increased by rhGUS therapy

BACKGROUND

Skeletal Disease in MPS VII

In MPS disorders, glycosaminoglycans accumulate in the lysosomes of cells in the joints and bones including chondrocytes, osteoblasts, and osteoclasts, which disrupts the normal cycle of bone formation and remodeling leading to abnormal bone structure and function, impaired growth, pain, and disability. Patients with MPS VII can develop a number of skeletal complications requiring multiple orthopedic surgeries and frequently become wheelchair bound by their mid-teens or early 20s.

Characteristic Skeletal abnormalities of MPS VII:

- Short stature
- Macrocephaly
- Atlanto-Axial instability
- Spinal cord compression
- Hip dysplasia
- Joint stiffness and arthropathy
- Kyphoscoliosis
- Gibbous deformity of spine
- Valgus deformity of knee
- Pectus Carinatum

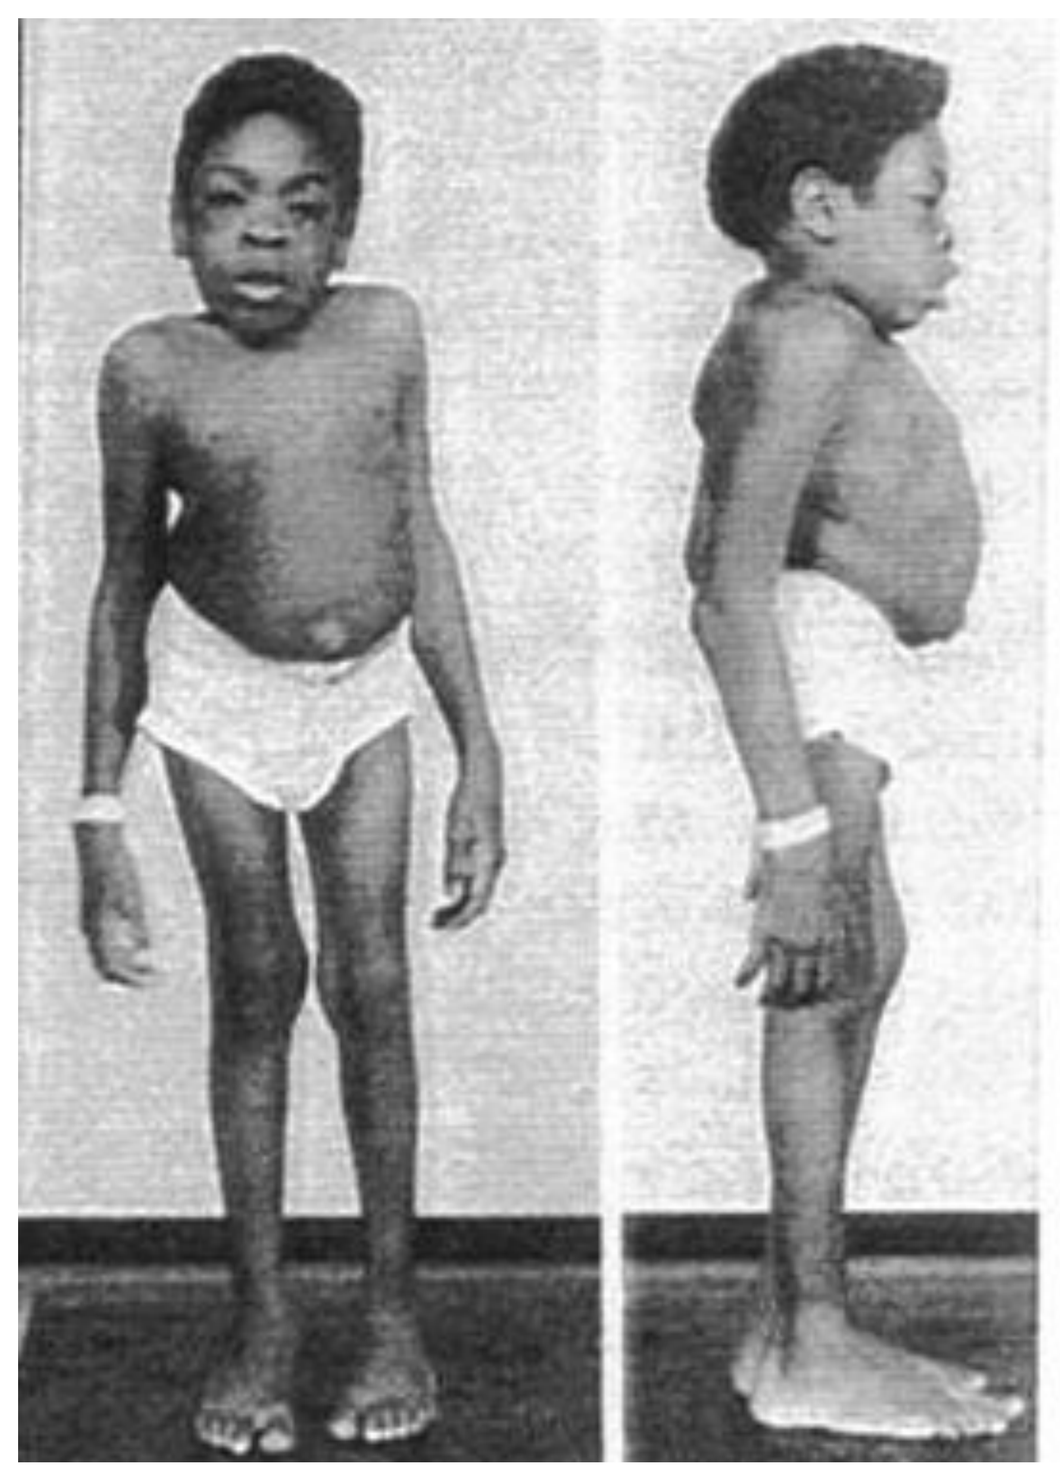

Enzyme replacement therapy has been demonstrated to improve cellular defects in bone remodeling in MPS animal models and skeletal manifestations have shown clinical response in patients with MPS disease including improved mobility (Hendriksz 2014; Tomatsu 2015, Rowan 2012.).

This is the first clinical report of changes in bone biomarkers after infusion of the investigational product rhGUS (UX003) in a child with MPS VII.

CLINICAL LABORATORY RESULTS

Table 1. Bone Biomarkers for 3 yo MPS VII Patient in Clinical Trial of rhGUS

Study Week	ALP (U/L) Ref range 104-345	BALP (U/L)[a] Ref range 31-103	P1NP (mcg/L) Ref range 22-105	CTX (pg/mL) Ref Range 1500-1700
Screening	186	------	------	------
Baseline[b]	139	------	------	------
Week 2	1633	------	------	------
Week 4	402	111	470	822
Week 8	185	------	------	------
Week 16	181	47.3	410	788

Abbreviations: ALP = alkaline phosphatase; BALP = bone-specific alkaline phosphatase; CTX = C-terminal telopeptide; P1NP = Procollagen Type 1 N-terminal propeptide
[a] BALP measured by Quantitative Chemiluminescent Immunoassay
[b]Baseline measurement taken before first infusion

CONCLUSIONS

- These early data in a single MPS VII pre-pubertal patient treated with rhGUS provide preliminary evidence that enzyme replacement therapy may stimulate bone formation.
- Based on historical studies in MPS animal models, the early initiation of enzyme replacement therapy can improve bone morphology consistent with the observed effect of rhGUS on bone turnover markers in this young patient (Sands et al 1994, Byers et al 1997).
- Bone formation biomarkers will be explored in other prepubertal patients with MPS VII in addition to monitoring growth while on long term treatment with rhGUS.

PATIENT DESCRIPTION

Presentation

- Full term infant born without complications to non-consanguinous parents; prenatal ultrasound at 20 wks gestation showed thickened nuchal fold but not seen on repeat ULS one week later; newborn exam unremarkable
 - Weight 3995 grams (75% CDC weight for age, boys birth-36 mos)
 - Length 50cm (50% CDC length for age, boys birth-36 mos)
 - Head Circumference 35.5 cm (50% CDC head circumference for age, boys birth-36 mos)
- Age 36 days, presented with petechiae of palate and arms; initial workup revealed thrombocytopenia requiring platelet transfusion; underwent course of IV immunoglobulin for presumed idiopathic thrombocytopenia; bone marrow biopsy revealed abnormal granulations in white blood cells.
- Initial urine glycosaminoglycans mildly elevated; screening MPS enzyme assay revealed low *β*-glucuronidase activity
- Genetic testing revealed compound heterozygosity for mutations in the *GUSB* gene: c.88C>T (p.P30S) (known mutation) and c.290 G>C (p.G97A) (predicted pathogenic variant)

Clinical Course

- Currently, a 42-month-old boy with short stature, failure to thrive, mild coarse facial features, with normal vision and hearing, and normal motor, cognitive and language development
- Growth deceleration in both length and weight noted at age 16 months (see growth chart)
- Normal ECHO; No recurrent otitis media
- Inguinal hernia diagnosed at age 12 months and repaired at age 26 months
- Abdominal ultrasound performed at age 35 months revealed liver at upper limits of normal and normal size spleen

Study Course

- Subject was consented at 35 months of age and screened for enrollment in UX003-CL203 to investigate the safety and efficacy of rhGUS
- Started on 4mg/kg of rhGUS every other week
- After initial infusion, noted that alkaline phosphatase was significantly elevated
- Additional bloodwork done to ensure that elevation was due to bone source rather than liver (see Table 1) with verification of normal hepatic transaminases, amylase, gamma glutamyl transferase, thyroid function, parathyroid function, and vitamin D-25-OH levels
- As of week 14, he has tolerated infusions without any associated reactions
- His current length as of age 40 months is 88.5 cm (+1.4cm since screening), weight 13.4 kg (+1.9 kg increase since screening, now 25% CDC weight for age, boys age 2 -20 yo), stable head circumference

PHYSIOLOGY OF BONE REMODELING

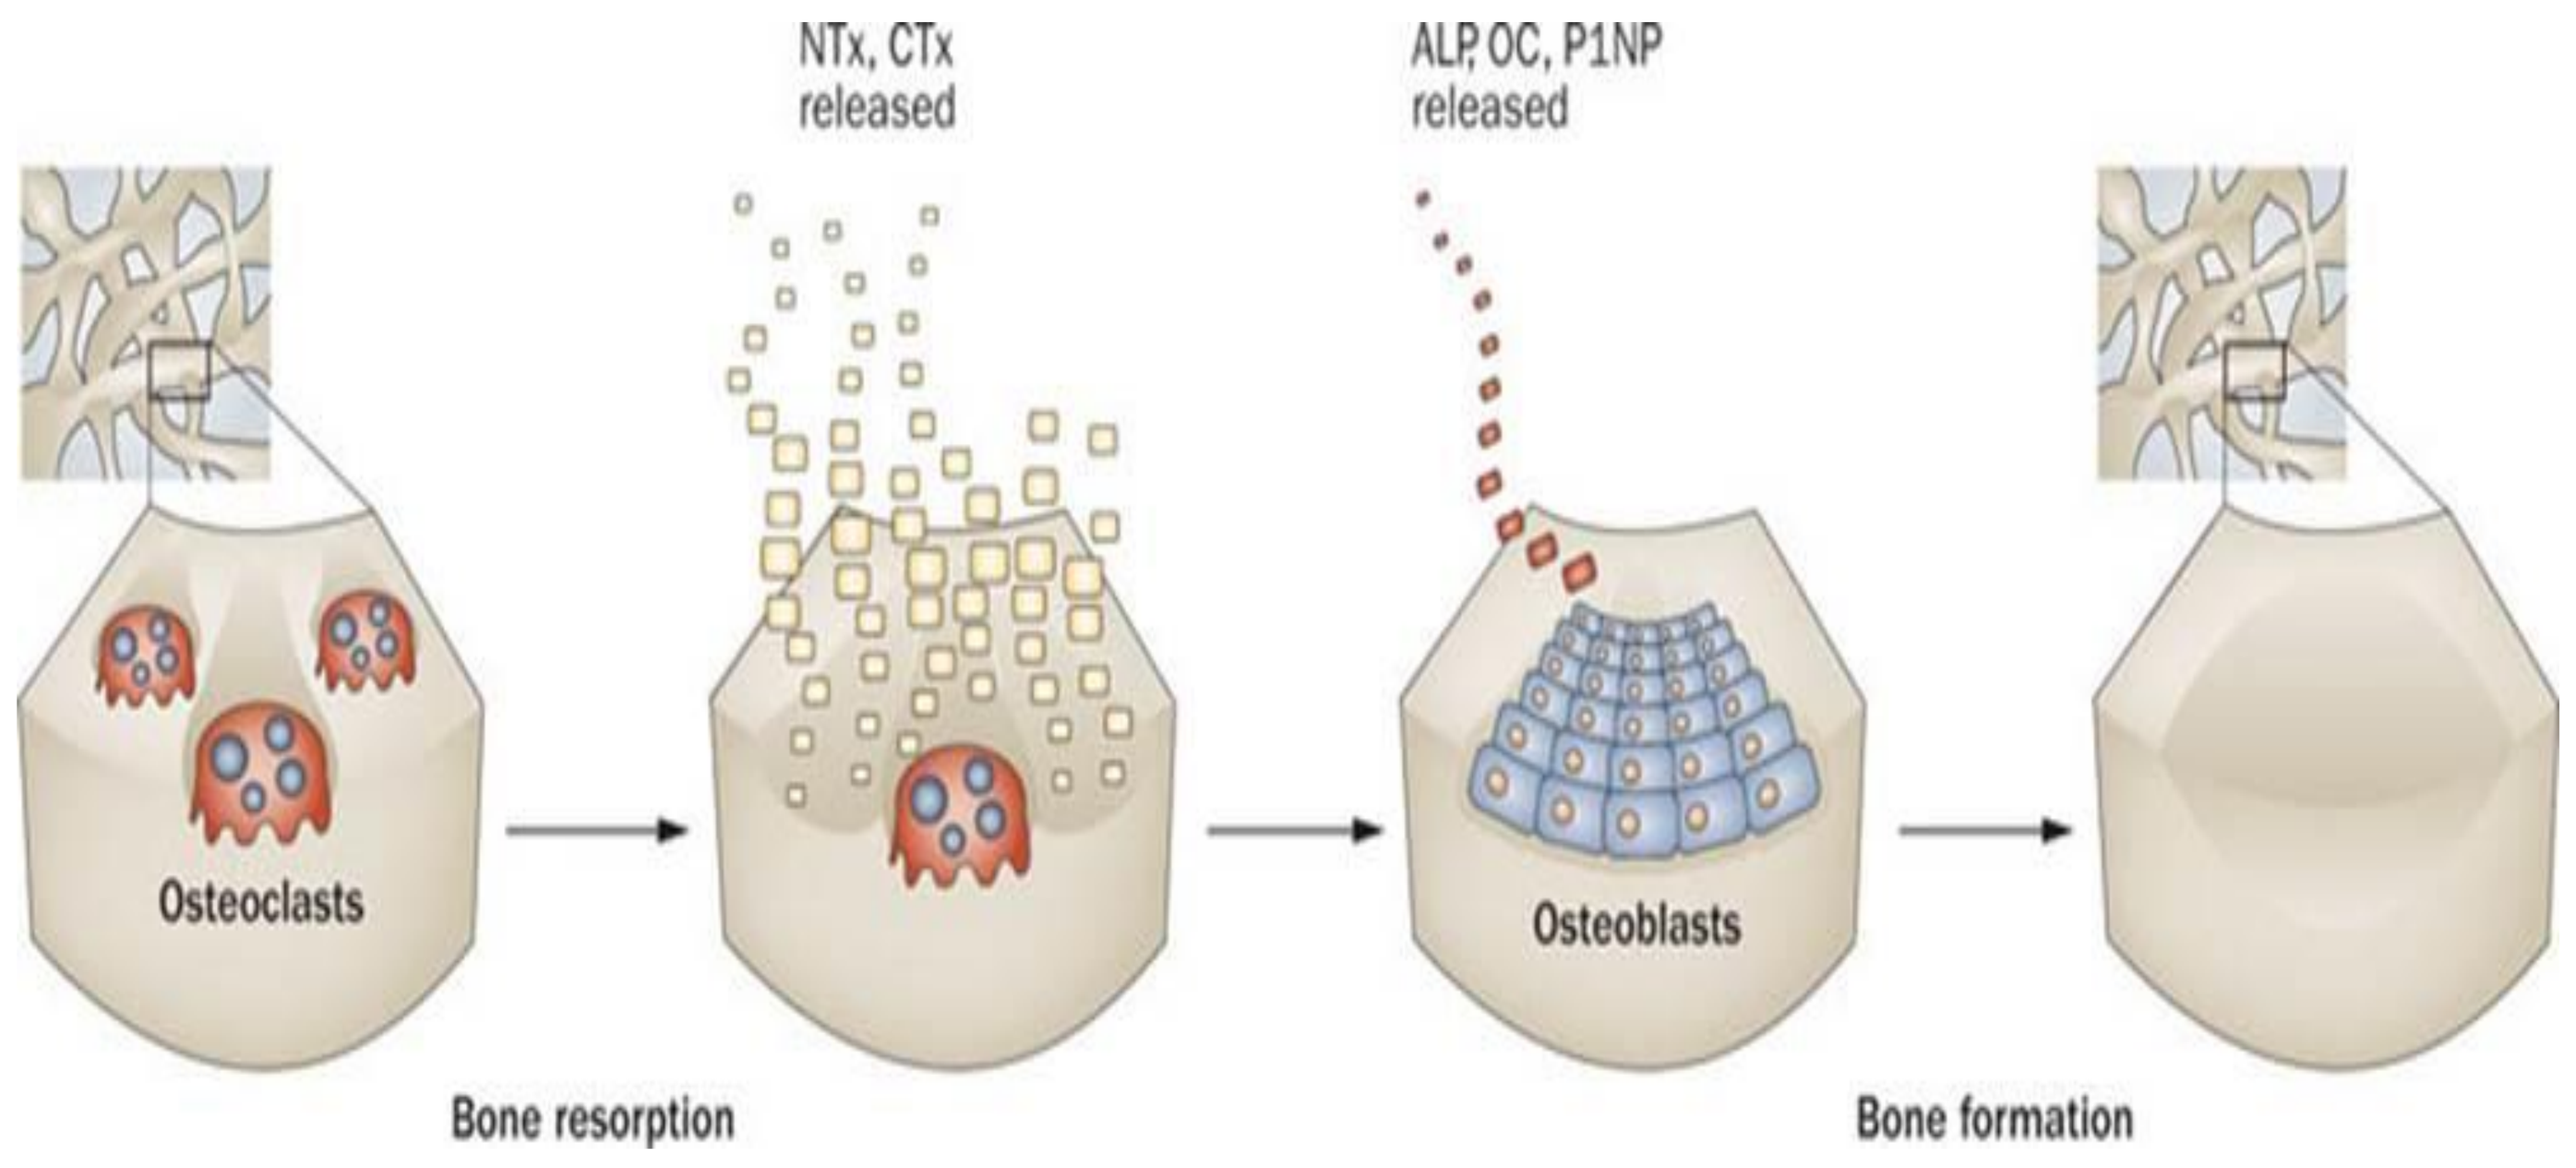

NTx, CTx released
ALP, OC, P1NP released
Osteoclasts
Osteoblasts
Bone resorption
Bone formation

Source: Suva et al. Nat Rev Endocrinol. 2011;7:208-218

REFERENCES

1. Kuehn SC et al. Impaired bone remodeling and its correction by combination therapy in a mouse model of mucopolysacchardiosis I. *Human Mol Gen.* 2015:7075-86.
2. Rowan DJ et al. Long circulating enzyme replacement therapy rescues bone pathology in mucopolysaccharidosis VII murine model. *Mol Genet Metab*. 2012;107:161-172.
3. Hendriksz CJ et al. Efficacy and safety of enzyme replacement therapy with BMN 110 (elosulfase alfa) for Morquio A syndrome (mucopolysaccharidosis IVA): a phase 3 randomised placebo-controlled study. *J Inherit Metab Dis.* 2014;37:979-990.
4. Tomatsu S et al. Therapies for the bone in mucopolysaccharidosis. *Mol Genet Metab*. 2015;114:94-109.
5. Herrmann D et al. Reference values of bone stiffness index and C-terminal telopeptide in healthy European children. *Int J Obes* (Lond). 2014;38(Suppl 2):S76-85.
6. Sands MS et al. Enzyme replacement therapy for murine mucopolysaccharidosis Type VII. *J Clin Invest*. 1994;93:2324-2331.
7. Byers S et al. Effect of enzyme replacement therapy on bone formation in a feline model of mucopolysaccharidosis type VI. *Bone*. 1997;425-431.